F2-875

Human recombinant antibody (Ab) cocktails protect against Methicillin Resistant *Staphylococcus aureus* (MRSA) infection in mice

Quinton Zondervan*, Kimberly Carey*, Laura Croal*, Maya Dubey*, Joseph Porter*, Robyn Puro*, Elizabeth Reczek*, Hugh Russell*, Rachel Schneider*, Theresa Towle*, Gwen Wilmes*, Yoram Puius†, Gail Skowron† and Vincent Coljee*

*Excelimmune, Inc, Woburn, MA, USA

†Roger Williams Medical Center, Providence, RI, USA

50 YEARS ICAAC

Boston ,MA

September 12-15th

Abstract (Revised)

Background: *Staphylococcus aureus* is a Gram-positive pathogen that causes disease through a diverse set of virulence factors and toxins. In the U.S., it now causes more deaths than HIV/AIDS and tuberculosis combined and is increasingly resistant to antibiotics. The synergistic activity of human recombinant antibody cocktails targeting multiple factors and toxins has the potential to enhance the efficacy of passive immunotherapy beyond that of monoclonal antibodies and to curtail the development of resistant strains of *S. aureus*.

Methods: Antibodies (Abs) are cloned from healthy human *S. aureus* carriers, expressed *in vitro* and screened for ability to bind *S. aureus* antigens. *S. aureus* binding Abs are evaluated and tested in a mouse *S. aureus* infection model for ability to enhance survival.

Results: At total antibody concentrations as low as 1 mg/kg (Ab/body weight), human recombinant Ab mixtures are fully protective in mice against lethal doses of MRSA. We demonstrate that a 5 antibody cocktail is capable of nearly doubling the LD_{50} dose of MRSA from 2.75 x 10^8 to 5.0 x 10^8 CFU.

Conclusions: Bacterial pathogens develop resistance to most known anti-microbial therapeutics over time. We describe a recombinant *S. aureus*-specific, fully human Ab therapy capable of protecting mice at lethal infection levels that may be immune to development of bacterial resistance. The approach is to develop Abs against a range of known and unknown antigens to design the most efficacious Ab cocktail representing the diversity of natural immunity found in healthy human donors.

Background

Antibiotic-resistant strains of *S. aureus* are on the rise and are a major health concern, both in the hospital setting and, increasingly, in the community at large. Methicillin-resistant *S. aureus* (MRSA) is currently a leading cause of death due to infectious disease in the United States, surpassing deaths from HIV/AIDS and tuberculosis combined [1, 2]. The death rate from invasive MRSA infection is now 20% [2]. Resistant forms of *S. aureus* have emerged within a few years of the introduction of each new class of antibiotics [3, 4]. Given the current status of research and development of new antibiotics, resistance in bacteria is increasing faster than our ability to develop novel antibiotics [5, 6]. Alternative treatments are needed to replace those that are increasingly ineffective.

By 1910, Meakins had already demonstrated that antibodies, as an enriched immunoglobulin, are protective against *S. aureus* infections in humans [7]. In addition, his research showed that this protection functioned against not only one strain, but against many. In the years since those results were published a century ago, it has become well established in the field of immunology that antibodies are capable of working synergistically to provide protection against *S. aureus* and to eliminate ongoing infection.

In 1962, Koenig et al. demonstrated that antibodies raised against *S. aureus* alpha toxin could protect animals from purified alpha toxin but not from infection with a replicating, toxigenic *S. aureus* strain [8]. In contrast, antibodies raised against heat killed *S. aureus* could protect against live challenge, but not against culture supernatants rich in alpha toxin. These results clearly demonstrated that approaches targeting a single aspect of a complex bacterial pathogen like *S. aureus* are unlikely to succeed.

More recently, research into the *S. aureus* exoproteome has demonstrated that few strains express the same spectrum of antigens, and even antigens expressed in common between strains may be expressed at widely varying levels and in dissimilar temporal expression patterns [9-11]. Recent research has shown that humans infected with *S. aureus* respond with diverse humoral immune reactions to the organism [12, 13]. The heterogeneous nature of *S. aureus* infection makes it paramount to approach treatment of this organism with a multi-pronged attack.

In Russia, anti-*S.aureus* immunoglobulins were used successfully from the 1960s to the mid 1990s to treat severe sepsis and infections due to antibiotic resistant *S. aureus* strains [14]. However, efforts to modernize passive immunotherapy approach through monoclonal antibody technology have been largely unsuccessful and there are currently no such therapies against *S. aureus* in the clinic [15].

Despite the demonstrated potential of passive immunotherapies, they remain vastly underutilized. We believe that this represents an extraordinary opportunity to bring a basic immunology concept, that of antibodies working synergistically to combat a pathogen, to the clinic. Excelimmune is dedicated to the development of human recombinant polyclonal antibody (HRPA) therapeutics for the treatment of human disease. **Here we present data from our proof-of-concept study that demonstrates the ability of an oligoclonal antibody test cocktail to completely protect mice at the LD_{50} dose in a mouse model of *S. aureus* infection.**

Methods

Healthy subjects were recruited by posters located throughout the Roger Williams Medical Center campus. Blood from qualified subjects was processed through a Ficoll gradient to isolate PBMCs. Antibody-secreting plasma cells, identified by cell surface markers, were single-cell sorted into 96-well plates and subjected to RT-PCR to amplify antibody genes (**Figure 1A**). Amplified products (**Figure 1B**) were cloned and expressed in mammalian cells in 96-well format for screening. Expressed antibodies were screened by ELISA to identify *S. aureus* -specific binders (**Figure 1C-E**). Positives were reconfirmed through further ELISA testing (**Figure 1F**), sequenced and expressed in a larger scale batch culture for *in vitro* testing and animal studies.

***In vivo* Studies:** *S. aureus*-binding antibodies were evaluated in mouse studies measuring survival. In these studies, mice were infected with methicillin resistant *S. aureus* strain, USA300, by intraperitoneal injection with a range of doses of *S. aureus* surrounding the LD_{50} dose of 2.75 x10^8 CFU. Immediately following infection, groups of 10 mice were treated by intra-peritoneal injection with PBS, 110 mg/kg vancomycin, or 1.0 mg/kg of a 5 antibody cocktail. Animals were observed for three days post-treatment and live/dead status was noted.

RESULTS

Figure 1:

A.

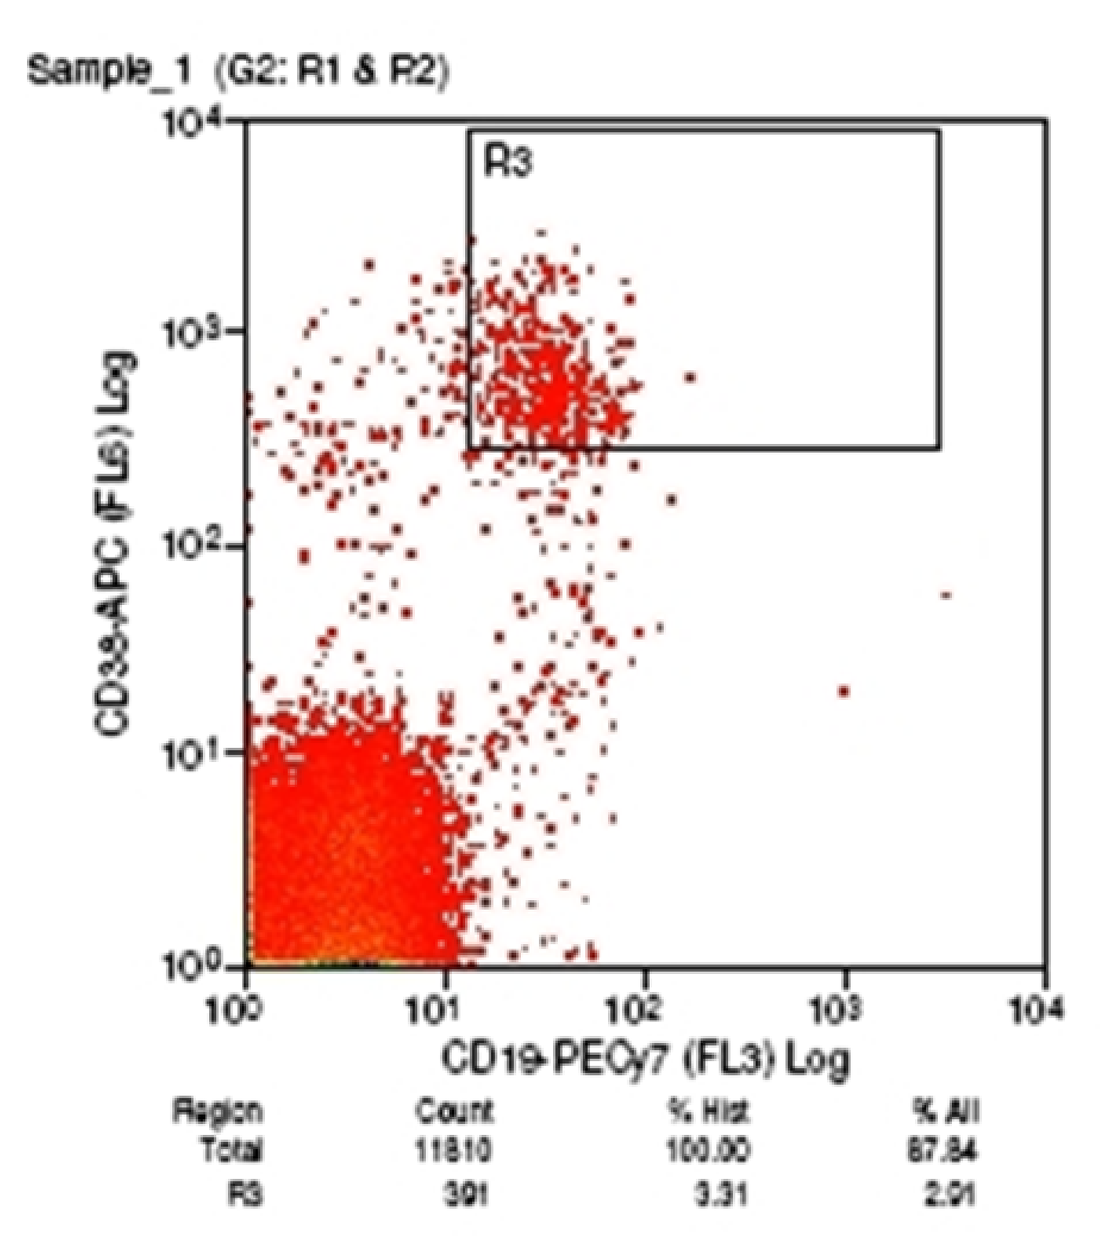

Sample_1 (G2: R1 & R2)
R3
CD38-APC (FL6) Log
CD19-PECy7 (FL3) Log
Region Count % Hist % All
Total 11810 100.00 87.84
R3 391 3.31 2.91

B.

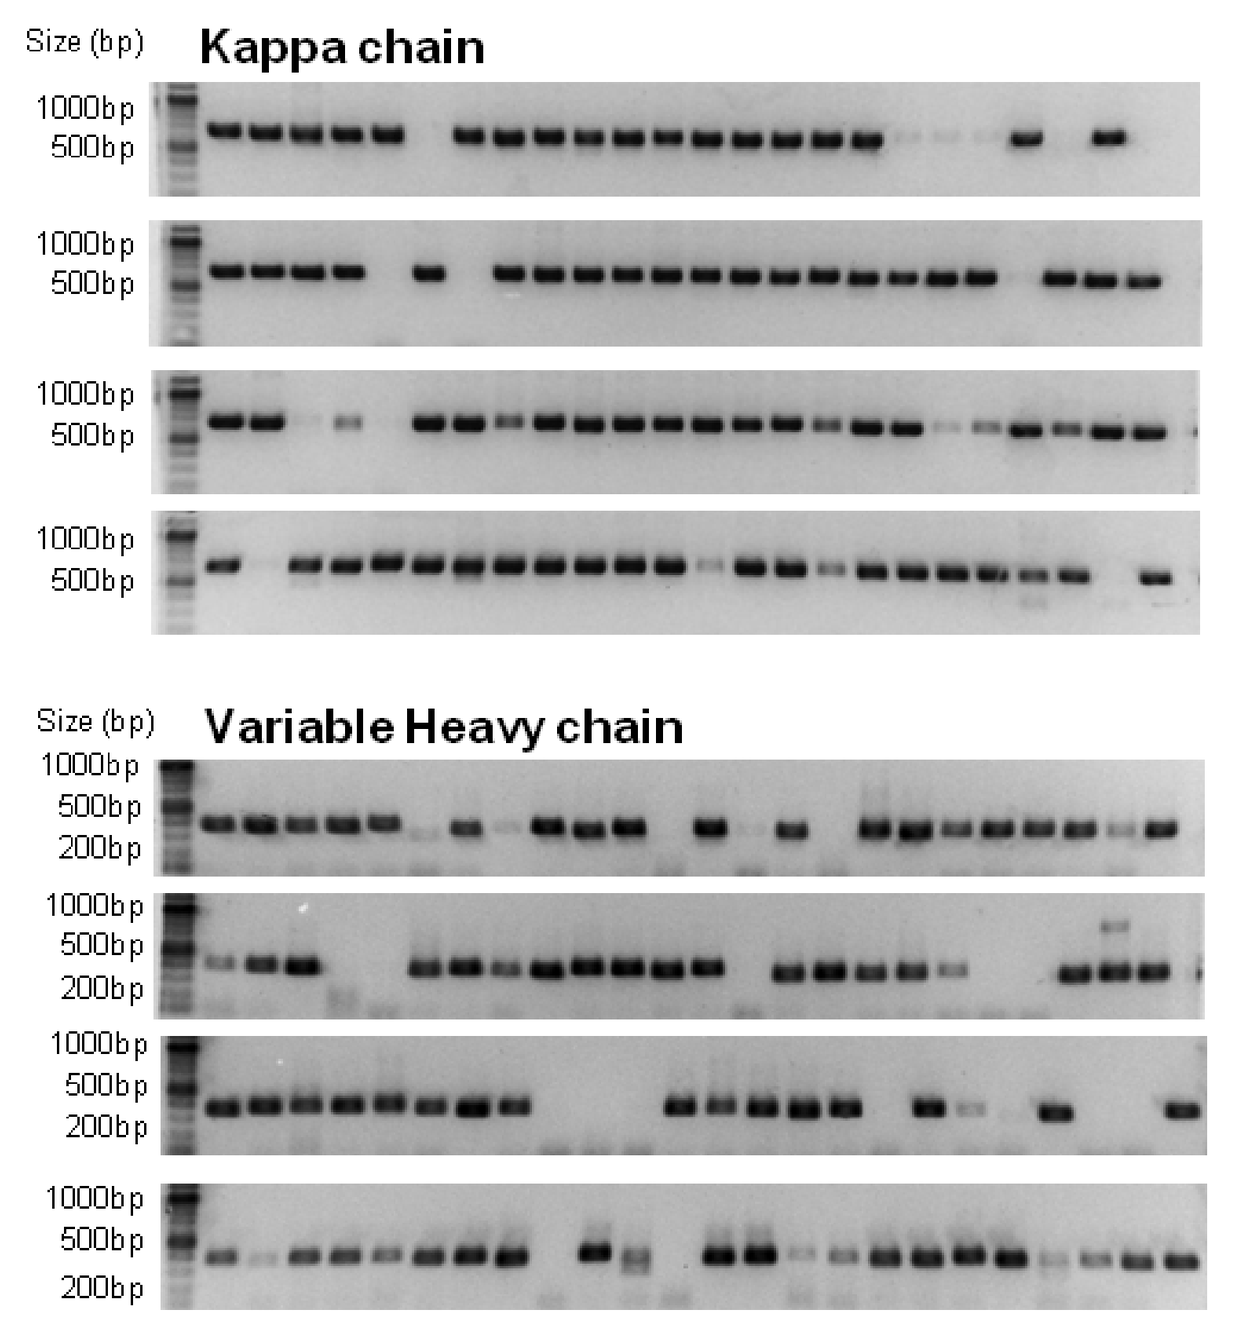

Size (bp)
Kappa chain
1000bp
500bp
Variable Heavy chain
200bp

C. Antibody expression ELISA

	1	2	3	4	5	6	7	8	9	10	11	12
A	1.096	1.477	1.725	0.259	0.230	0.089	0.107	0.493	1.735	0.093	1.540	1.903
B	0.699	1.555	0.085	1.910	0.139	1.865	1.877	2.056	0.078	0.351	0.083	0.097
C	0.990	1.100	1.833	0.175	1.688	0.107	2.290	1.858	0.198	1.974	0.416	1.662
D	1.473	0.392	1.832	1.405	1.666	1.776	1.732	1.692	0.940	0.161	1.258	0.100
E	1.696	0.082	0.080	1.968	1.561	2.090	1.866	0.527	1.851	0.153	1.410	1.641
F	0.179	0.088	1.490	0.107	0.169	1.504	0.181	0.201	1.652	0.196	0.744	1.385
G	1.268	1.352	1.365	1.216	1.115	1.019	0.087	1.301	1.520	1.842	1.735	0.103
H	0.086	1.165	1.371	1.297	0.834	1.378	1.468	0.326	1.162	0.123	1.687	1.387

0-0.166 · 0.167-0.500 · 0.501-1.000 · 1.001-1.500 · 1.501-2.000 · 2.001-2.500 · 2.501-3.000 · Positive control well

D. *S. aureus* ELISA condition #1

	1	2	3	4	5	6	7	8	9	10	11	12
A	0.080	0.091	0.074	0.080	0.083	0.075	0.076	0.086	0.073	0.079	0.105	0.075
B	0.095	0.082	0.077	0.073	0.072	0.086	0.072	0.098	0.072	0.078	0.076	0.077
C	0.095	0.077	1.115	0.079	0.079	0.076	0.085	0.076	0.074	0.078	0.625	0.101
D	0.120	0.089	0.075	0.119	0.074	0.075	0.078	1.113	0.082	0.074	0.074	0.078
E	0.097	0.075	0.074	0.073	0.072	0.073	0.084	0.083	0.075	0.073	0.075	0.109
F	0.107	0.092	0.079	0.081	0.076	0.070	0.074	0.091	0.074	0.071	0.081	0.104
G	0.107	0.076	0.079	0.071	0.082	0.074	0.068	0.073	0.073	0.075	0.076	0.101
H	0.120	0.099	0.103	0.101	0.099	0.092	0.106	0.114	0.106	0.107	0.145	0.125

Above 2X background · Threshold (2X background) = 0.160 · Negative control well · Positive control well

E. *S. aureus* ELISA condition #2

	1	2	3	4	5	6	7	8	9	10	11	12
A	0.086	0.077	0.076	0.086	0.080	0.080	0.094	0.092	0.107	0.114	0.080	0.103
B	0.075	0.070	0.066	0.067	0.072	0.069	0.068	0.069	0.070	0.072	0.076	0.094
C	0.072	0.071	0.104	0.070	0.067	0.066	0.071	0.070	0.067	0.069	0.086	0.104
D	0.074	0.070	0.069	0.069	0.072	0.074	0.068	0.635	0.067	0.078	0.072	0.102
E	0.074	0.069	0.067	0.083	0.076	0.070	0.081	0.073	0.066	0.075	0.078	0.107
F	0.082	0.072	0.068	0.073	0.076	0.070	0.072	0.074	0.074	0.075	0.077	0.095
G	0.088	0.073	0.073	0.081	0.076	0.073	0.077	0.074	0.071	0.083	0.089	0.102
H	0.081	0.089	0.084	0.083	0.085	0.091	0.083	0.087	0.076	0.081	0.092	0.096

Above 2X background · Threshold (2X background) = 0.145 · Negative control well

F. Confirmation of Putative Positives

	1	2	3	4	5	6	7	8	9	10	11	12
A	0.060	0.061	0.058	0.061	0.060	0.058	0.058	0.058	0.059	0.056	0.067	0.057
B	0.063	0.229	0.070	0.099	0.070	1.379	0.065	0.065	0.068	0.068	0.059	0.060
C	0.059	0.209	0.067	0.098	0.063	1.351	0.066	0.068	0.064	0.064	0.060	0.060
D	0.062	0.058	0.062	0.058	0.061	1.434	0.066	0.065	0.066	0.065	0.061	0.063
E	0.060	0.060	0.059	0.059	0.059	0.059	0.059	0.059	0.059	0.059	0.059	0.060
F	0.057	0.196	0.059	0.082	0.057	1.357	0.062	0.056	0.059	0.061	0.055	0.053
G	0.059	0.197	0.059	0.084	0.058	1.270	0.061	0.055	0.059	0.059	0.052	0.054
H	0.058	0.053	0.053	0.052	0.054	0.053	0.053	0.053	0.053	0.052	0.050	0.052

Above 2X background · Threshold (2X background) = 0.129 · Negative control well · Positive control well

Figure 1: Data from a representative plate of fully human antibodies isolated through the ExcelAb antibody cloning and screening platform.
Panel A shows the population of CD38+/CD19+ plasma cells targeted for cloning. **Panel B** shows the amplified variable heavy and light antibody genes from individual plasma cells in a 96-well plate. Note the variation in size between individual amplicons in the heavy chain gel, which suggest amplification of a single heavy chain per well. Sequence data from cloned antibodies supports this conclusion. **Panel C** shows representative data from an antibody expression ELISA showing that the majority of amplified antibody cognate pairs are successfully cloned and are able to express antibody. **Panels D and E** show data from screening by ELISA in which the "hit" recognized *S.aureus* proteins in two separate ELISA conditions. The hit was confirmed with multiple replicates in the repeat ELISA shown in **Panel F**.

Figure 2: Human recombinant antibody cocktails protect against live *S. aureus* challenge in mice. An IP dose of 2.75 x10^8 CFU *S. aureus* strain USA300 was identified as the LD_{50} dose, sufficient to kill 50% of animals treated with PBS. Treatment with a cocktail composed of 5 human recombinant antibodies at a total antibody concentration of 1 mg/kg (Ab/body weight), was fully protective against the LD_{50} dose of MRSA. At the 3.5x10^8 CFU dose, all of the mice treated with a 5 antibody cocktail survived the full 72 hour study. At 5x10^8 CFU, or nearly twice the LD_{50} dose for PBS treated animals, 50% of the mice treated with the 5 antibody cocktail survived the full 72 hour study. Vancomycin, rescued all mice. All groups contained 10 mice.

Figure 3: Treatment with human recombinant antibody cocktails significantly shifts the LD_{50} curve for *S. aureus*. Mice treated with the 5 antibody cocktail were fully protected from a dose of 3.5x10^8 CFU, and displayed 50% mortality at a dose of 5x10^8 CFU. This represents a shift in the LD_{50} curve from 2.75x10^8 CFU to 5x10^8 CFU with the 5 antibody cocktail. A similar magnitude effect was observed in another study using a different *S. aureus* strain, although the LD_{50} for the two strains was significantly different.

Figure 2:

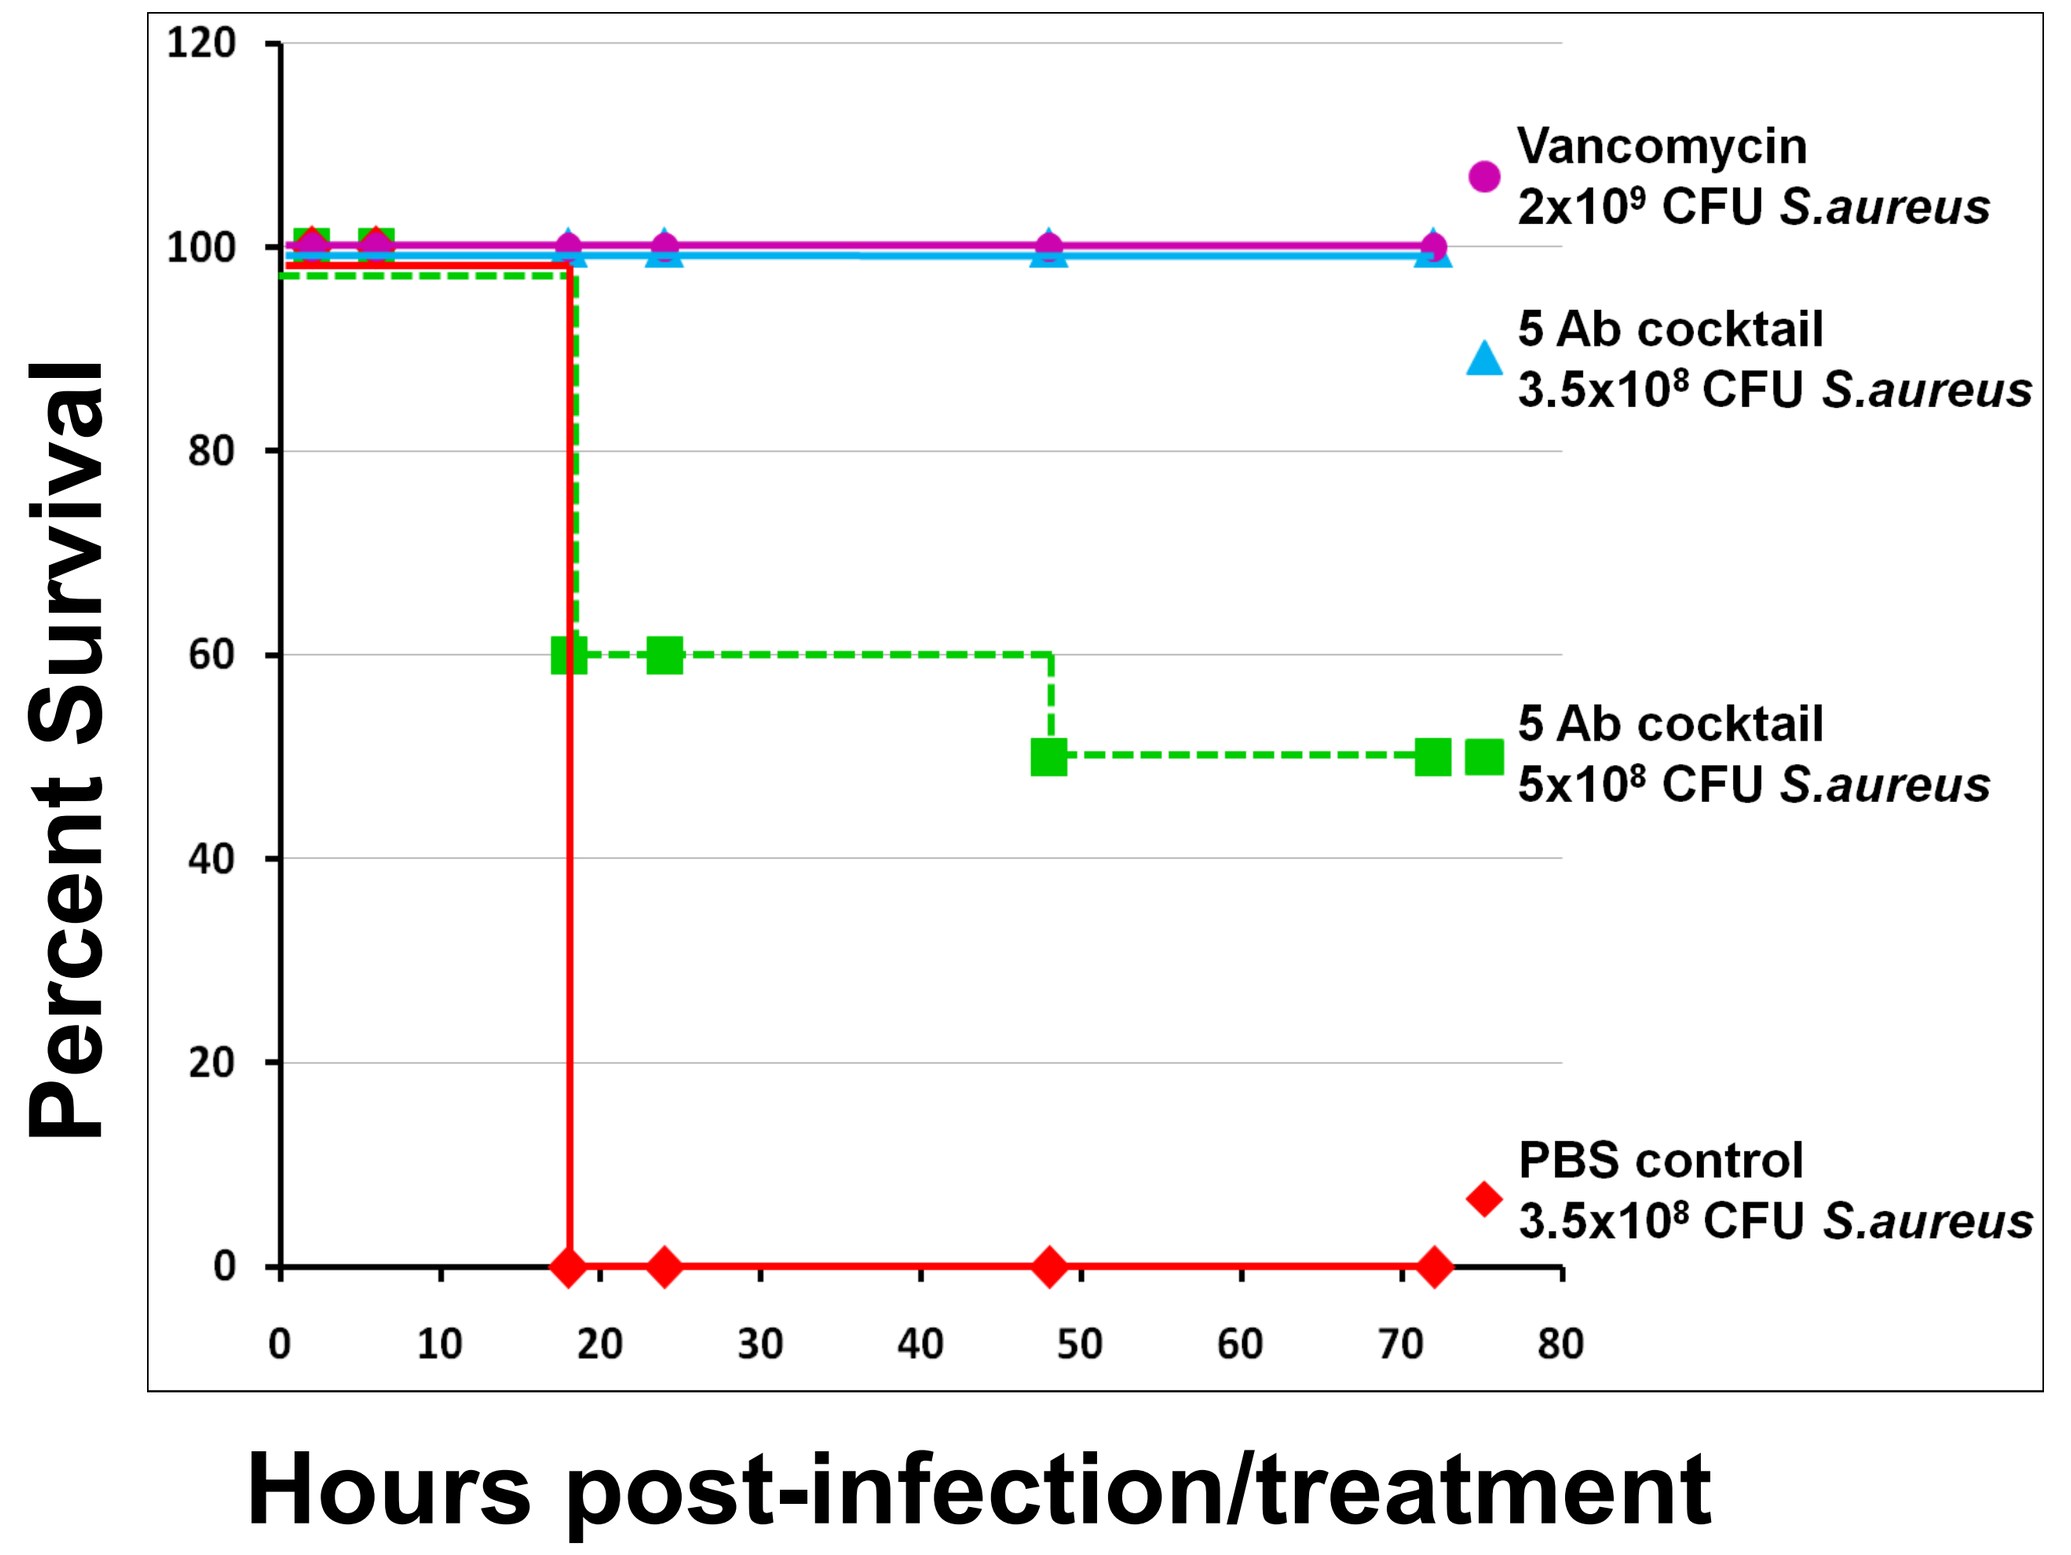

Percent Survival
Vancomycin
2x10⁹ CFU S.aureus
5 Ab cocktail
3.5x10⁸ CFU S.aureus
5 Ab cocktail
5x10⁸ CFU S.aureus
PBS control
3.5x10⁸ CFU S.aureus
Hours post-infection/treatment

Figure 3:

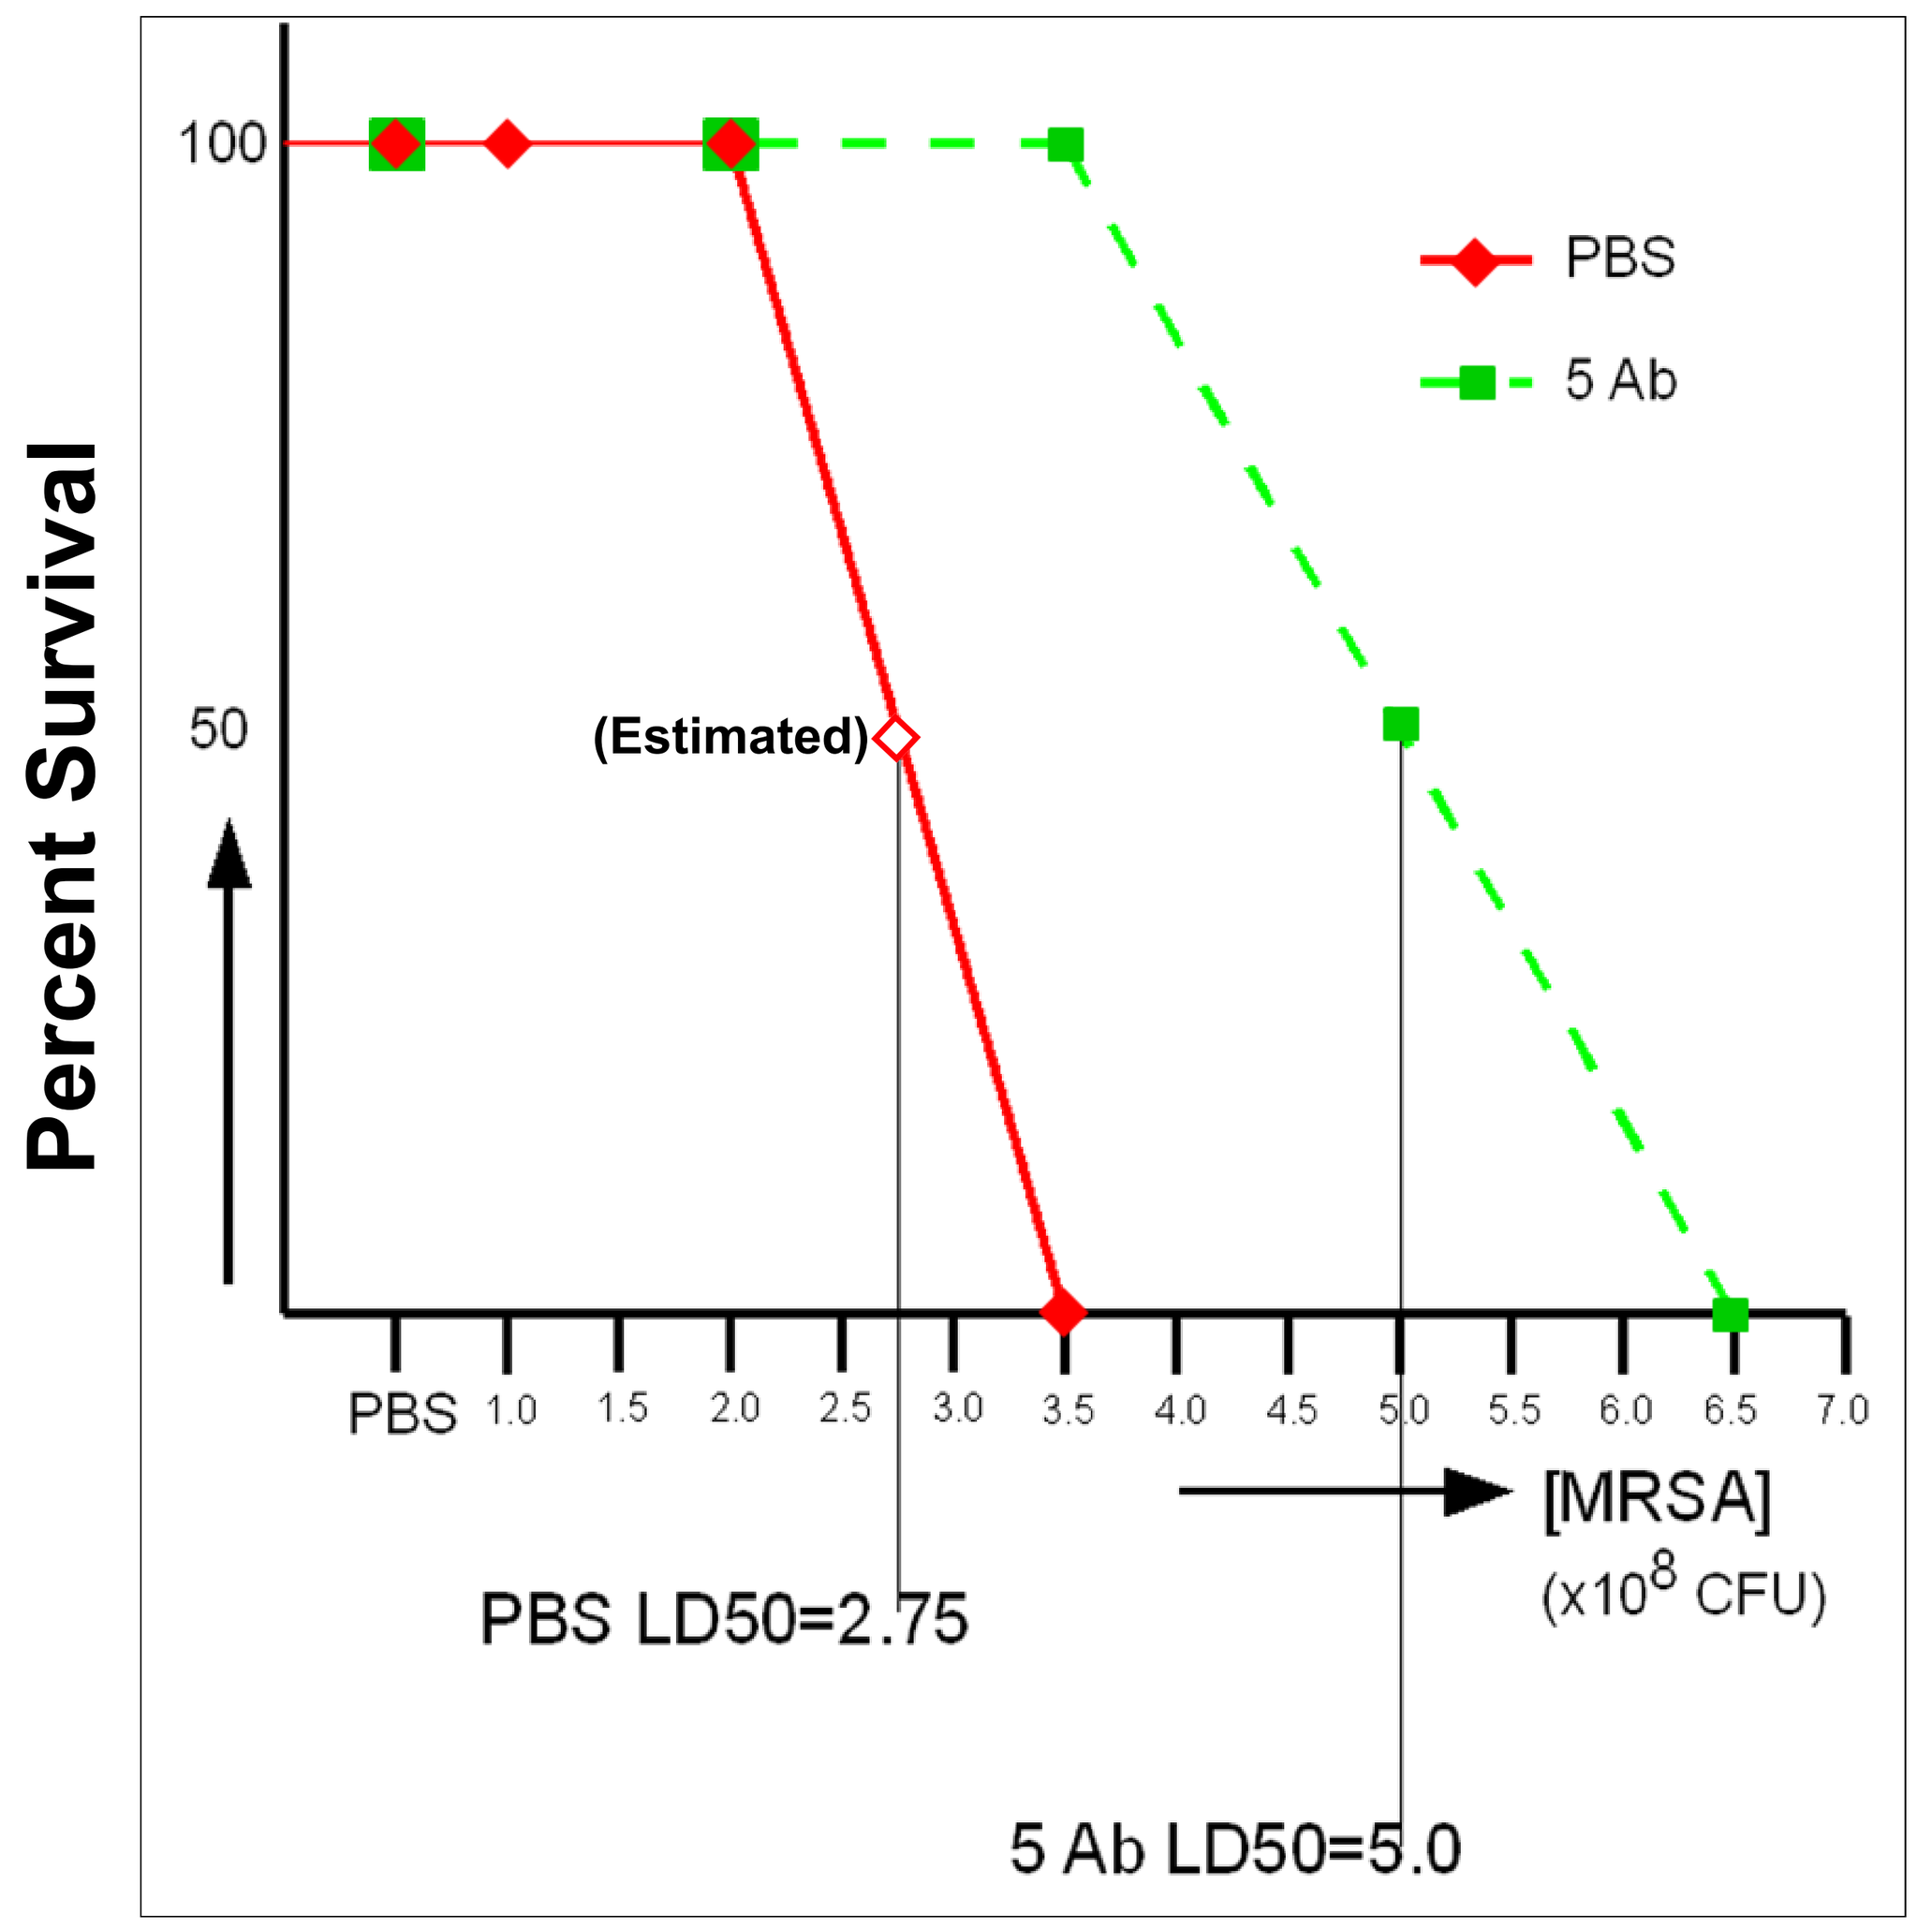

Percent Survival
PBS
5 Ab
(Estimated)
[MRSA]
(x10⁸ CFU)
PBS LD50=2.75
5 Ab LD50=5.0

Summary

- *S. aureus* is a highly variable organism capable of causing infections ranging in severity from mild to life threatening.
- Antibiotic resistance is on the rise both in the hospital and in community settings and the time required to develop a new antibiotic is far longer than the time required for resistance to develop.
- Monoclonal antibody approaches to bacterial infectious disease treatment are of limited utility due to the complex and diverse nature of bacterial pathogens. Much like antibiotics, monoclonal antibodies are susceptible to the development of resistant strains due to the monovalent nature of their mode of action.
- Recombinant polyclonal antibodies mimic the natural human immune response and are capable of providing protection against a wide range of virulence factors and bacterial strains with a greatly reduced chance of selection for resistant organisms.
- The synergistic action of the component antibodies in an Ab cocktail allows it to be effective at much lower doses than is possible with a conventional monoclonal antibody.
- Excelimmune has recently completed proof of concept studies demonstrating full protection at the LD50 dose of *S. aureus* in a mouse live bacterial challenge model. This effect was achieved at a dose far lower than that typically required for monoclonal antibodies.

References

1. Heron, M.P., Hoyert, D.L., Xu, J., Scott, C., and Tejada-Vera, B. , Deaths: preliminary data for 2006. . National vital statistics reports., 2008. 56: p. 1-52.
2. Klevens, R.M., et al., Invasive methicillin-resistant Staphylococcus aureus infections in the United States. JAMA, 2007. 298(15): p. 1763-71.
3. Boucher, H.W., et al., Bad bugs, no drugs: no ESKAPE! An update from the Infectious Diseases Society of America. Clin Infect Dis, 2009. 48(1): p. 1-12.
4. Kirby, W.M., Extraction of a Highly Potent Penicillin Inactivator from Penicillin Resistant Staphylococci. Science, 1944. 99(2579): p. 452-453.
5. Boucher, H.W. and G. Sakoulas, Perspectives on Daptomycin resistance, with emphasis on resistance in Staphylococcus aureus. Clin Infect Dis, 2007. 45(5): p. 601-8.
6. Walsh, C., Where will new antibiotics come from? Nat Rev Microbiol, 2003. 1(1): p. 65-70.
7. Meakins, J.C., An Experimental Study of Opsonic Immunity to Staphylococcus Aureus. J Exp Med, 1910. 12(1): p. 67-81.
8. Koenig, M.G., M.A. Melly, and D.E. Rogers, Factors relating to the virulence of Staphylococci. III. Antibacterial versus antioxic immunity. J Exp Med, 1962. 116: p. 601-10.
9. Bania, J., et al., The profiles of enterotoxin genes in Staphylococcus aureus from nasal carriers. Lett Appl Microbiol, 2006. 42(4): p. 315-20.
10. von Eiff, C., et al., Prevalence of genes encoding for members of the staphylococcal leukotoxin family among clinical isolates of Staphylococcus aureus. Diagn Microbiol Infect Dis, 2004. 49(3): p. 157-62.
11. Ziebandt, A.K., et al., Proteomics uncovers extreme heterogeneity in the Staphylococcus aureus exoproteome due to genomic plasticity and variant gene regulation. Proteomics, 2010.
12. Verkaik, N.J., et al., Heterogeneity of the humoral immune response following Staphylococcus aureus bacteremia. Eur J Clin Microbiol Infect Dis. 29(5): p. 509-18.
13. Verkaik, N.J., et al., Immunogenicity of toxins during Staphylococcus aureus infection. Clin Infect Dis. 50(1): p. 61-8.
14. Kelly, J., Immunotherapy against antibiotic-resistant bacteria: the Russian experience with an antistaphylococcal hyperimmune plasma and immunoglobulin. Microbes and Infection, 2000. 2: p. 1383-1392.
15. Garcia-Lara, J. and S.J. Foster, Anti-Staphylococcus aureus immunotherapy: current status and prospects. Curr Opin Pharmacol, 2009. 9(5): p. 552-7.